Trends in Specialty Drug Coverage Policy Complexity Among US Commercial Health Plans

Rucker JA[1], Enright DE[1], Lin Y[1], Beinfeld MT[1], Chambers JD[1]

1. Center for the Evaluation of Value and Risk in Health, Tufts Medical Center, Boston, MA

BACKGROUND & OBJECTIVE

- Health plan coverage policies influence patient access to specialty drugs. Complex coverage policies may increase administrative burden, delay treatment initiation, and discourage clinicians from prescribing specialty drugs.[1,2]
- Our study evaluates how the complexity of specialty drug coverage policies has evolved over time.

METHODS

Data Source

- We used coverage information from the Tufts Medical Center Specialty Drug Evidence and Coverage (SPEC) Database, which tracks specialty drug coverage decisions from the largest US commercial health plans.
- To ensure comparability across years, we restricted the analysis to the same 360 drug–indication pairs and the same 16 health plans included in SPEC each year from 2017–2025.

Analyses

- We assessed health plan coverage complexity overall and at the health plan level using four measures:

1. ***Policy length**:* defined as the word count of text describing the eligible patient population.
2. ***Number of coverage criteria:*** defined as the count of separately listed requirements in the eligibility text (e.g., age limits, diagnostic criteria).
3. ***Number of documents per coverage policy:*** reflecting cases in which health plans issued multiple documents to define access requirements.
4. ***Pre-authorization documentation requirements:*** indicating whether plans required submission of specific documentation prior to treatment approval (e.g., confirmation of impaired physical functioning).

- The analysis included approximately 4,550 specialty drug coverage policies annually, with minor year-to-year variation

RESULTS

- Between 2017 and 2025:
 - Mean policy length increased 169.5%
 - Mean number of individual coverage criteria increased 166.2%.
 - The share of coverage policies with multiple documents increased 32%
 - The share of coverage policies with documentation requirements increased 20.7%
- Stratified analysis by ICD-10 diagnostic category, orphan designation, and oncology status showed consistent trends.

Figure 1. Coverage Policy Mean Word Count & Number of Criteria (2017-2025)

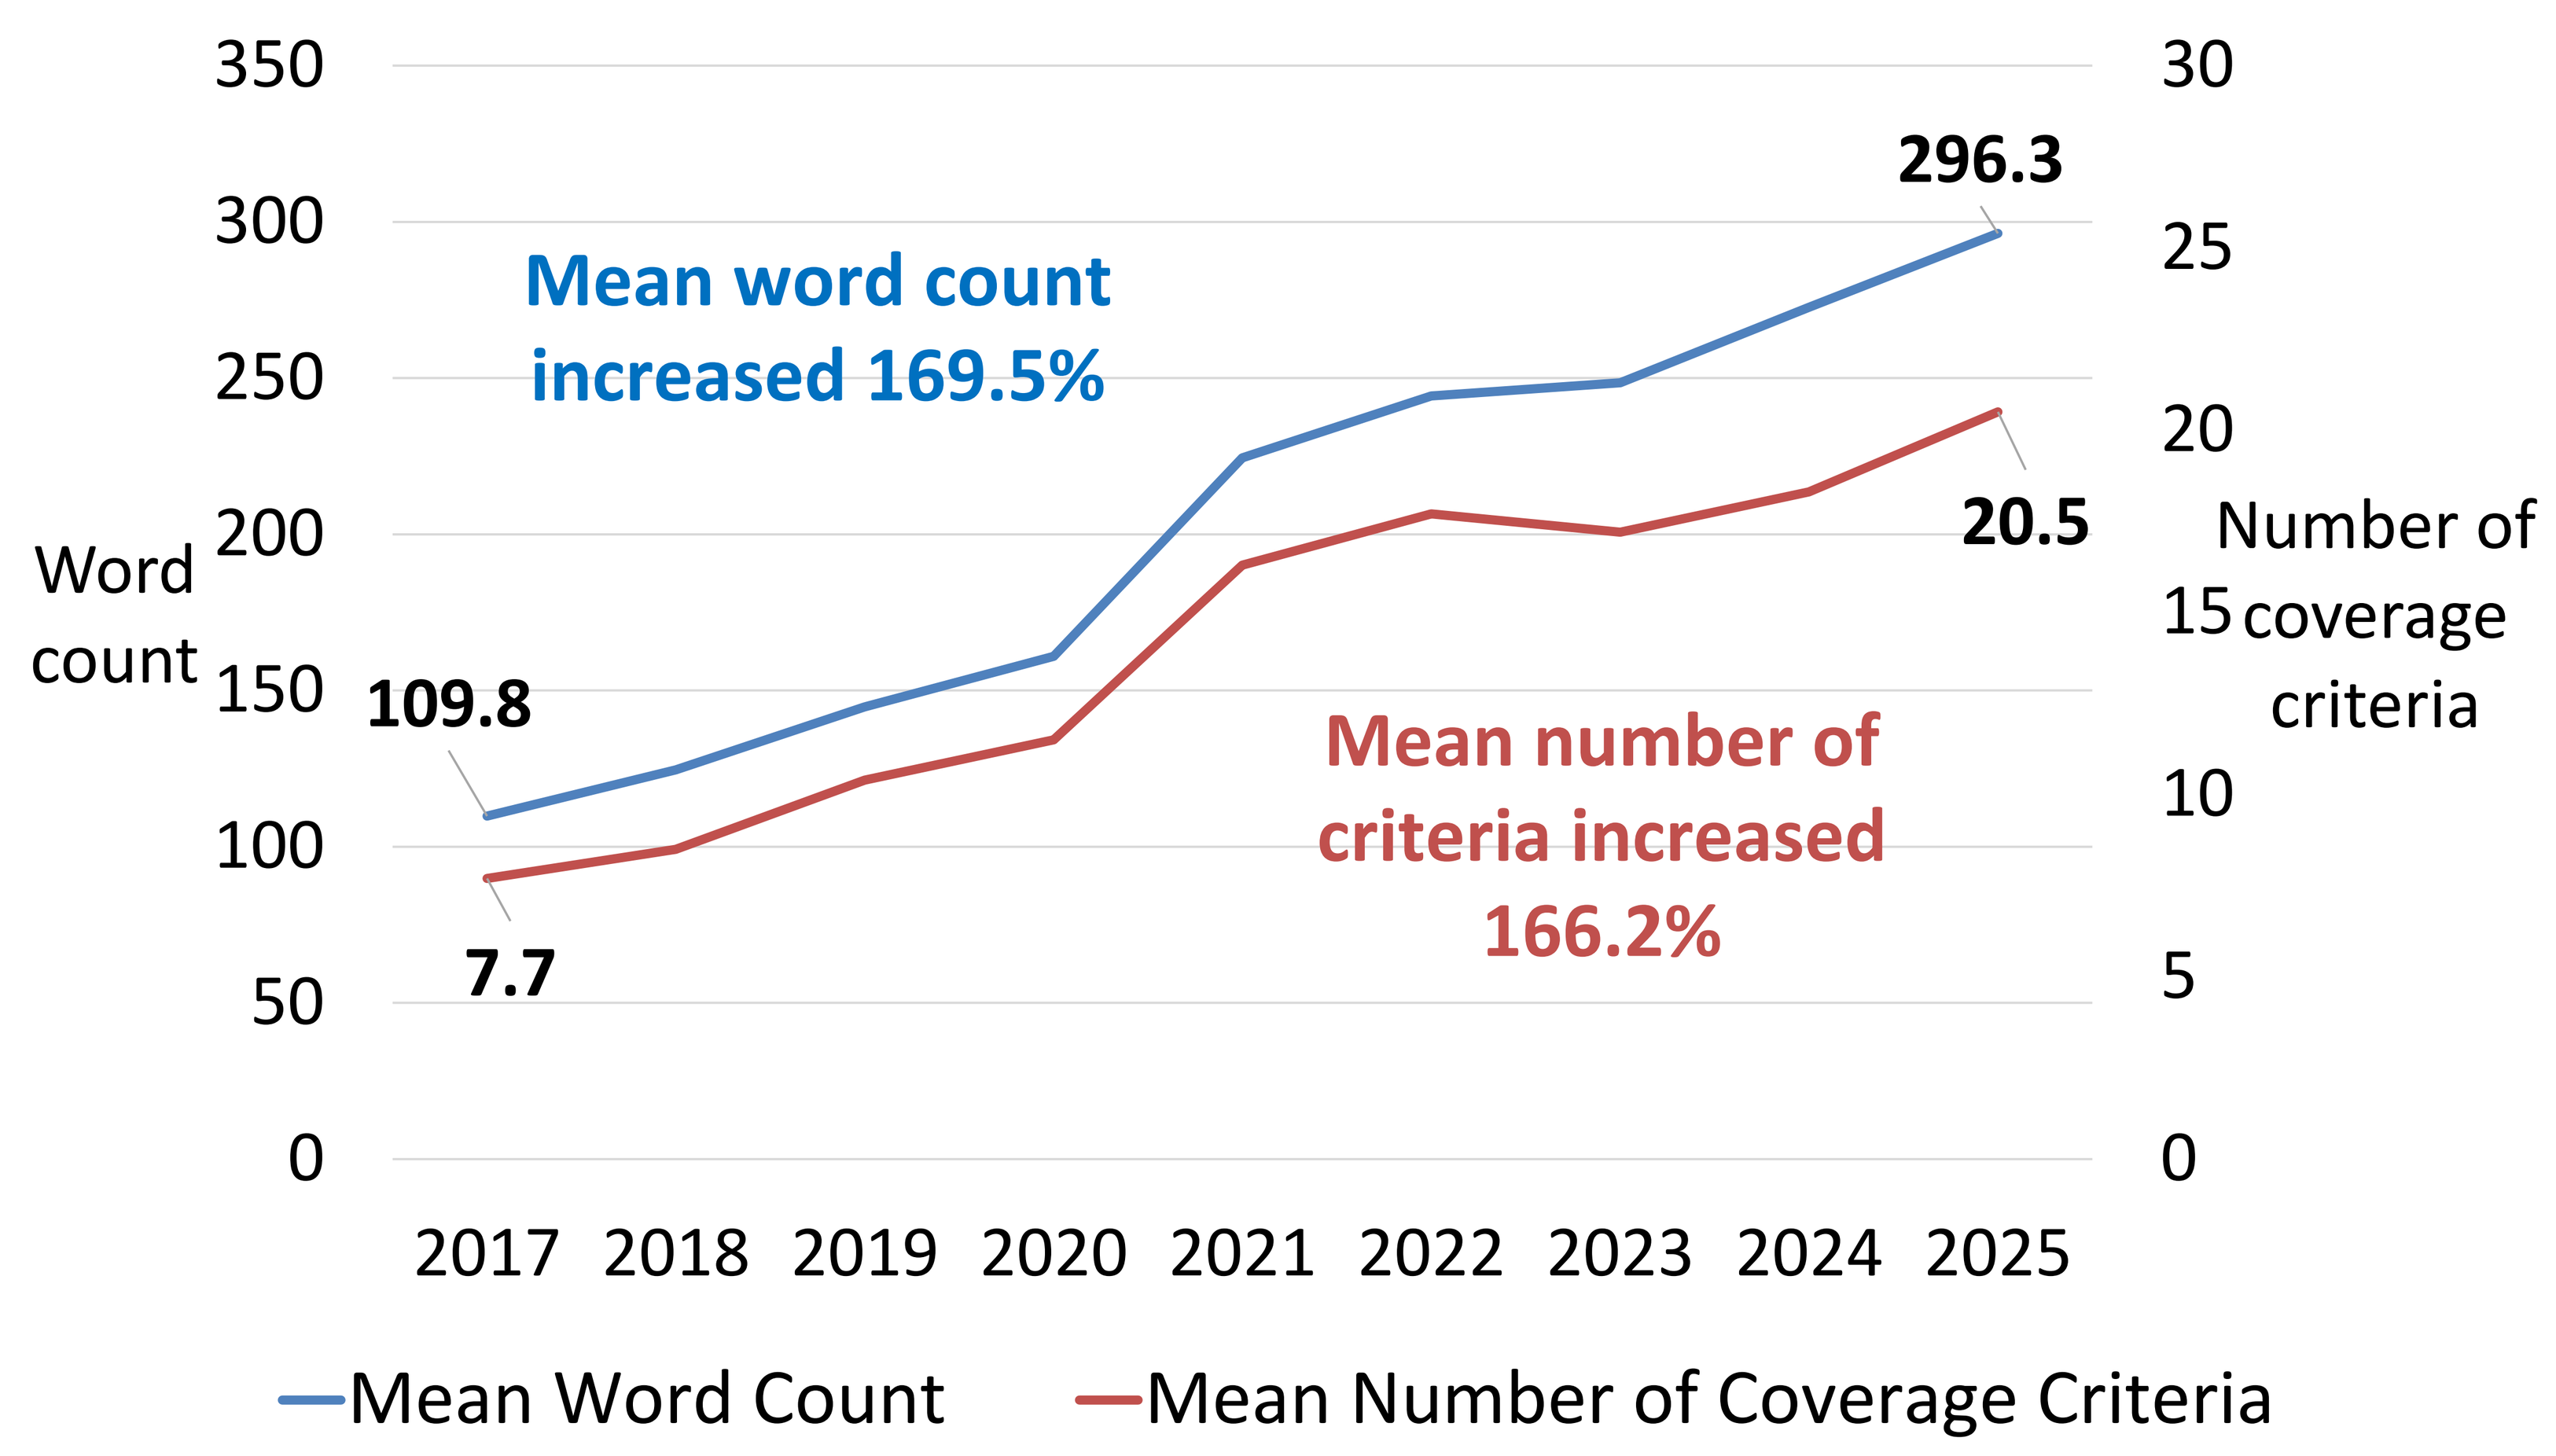
350
300
250
200
150
100
50
0
30
25
20
15
10
5
0
Word count
Number of coverage criteria
296.3
20.5
109.8
7.7
Mean word count increased 169.5%
Mean number of criteria increased 166.2%
2017 2018 2019 2020 2021 2022 2023 2024 2025
Mean Word Count
Mean Number of Coverage Criteria

Figure 2. Proportion of Coverage Policies with Multiple Documents and Documentation Requirements (2017-2025)

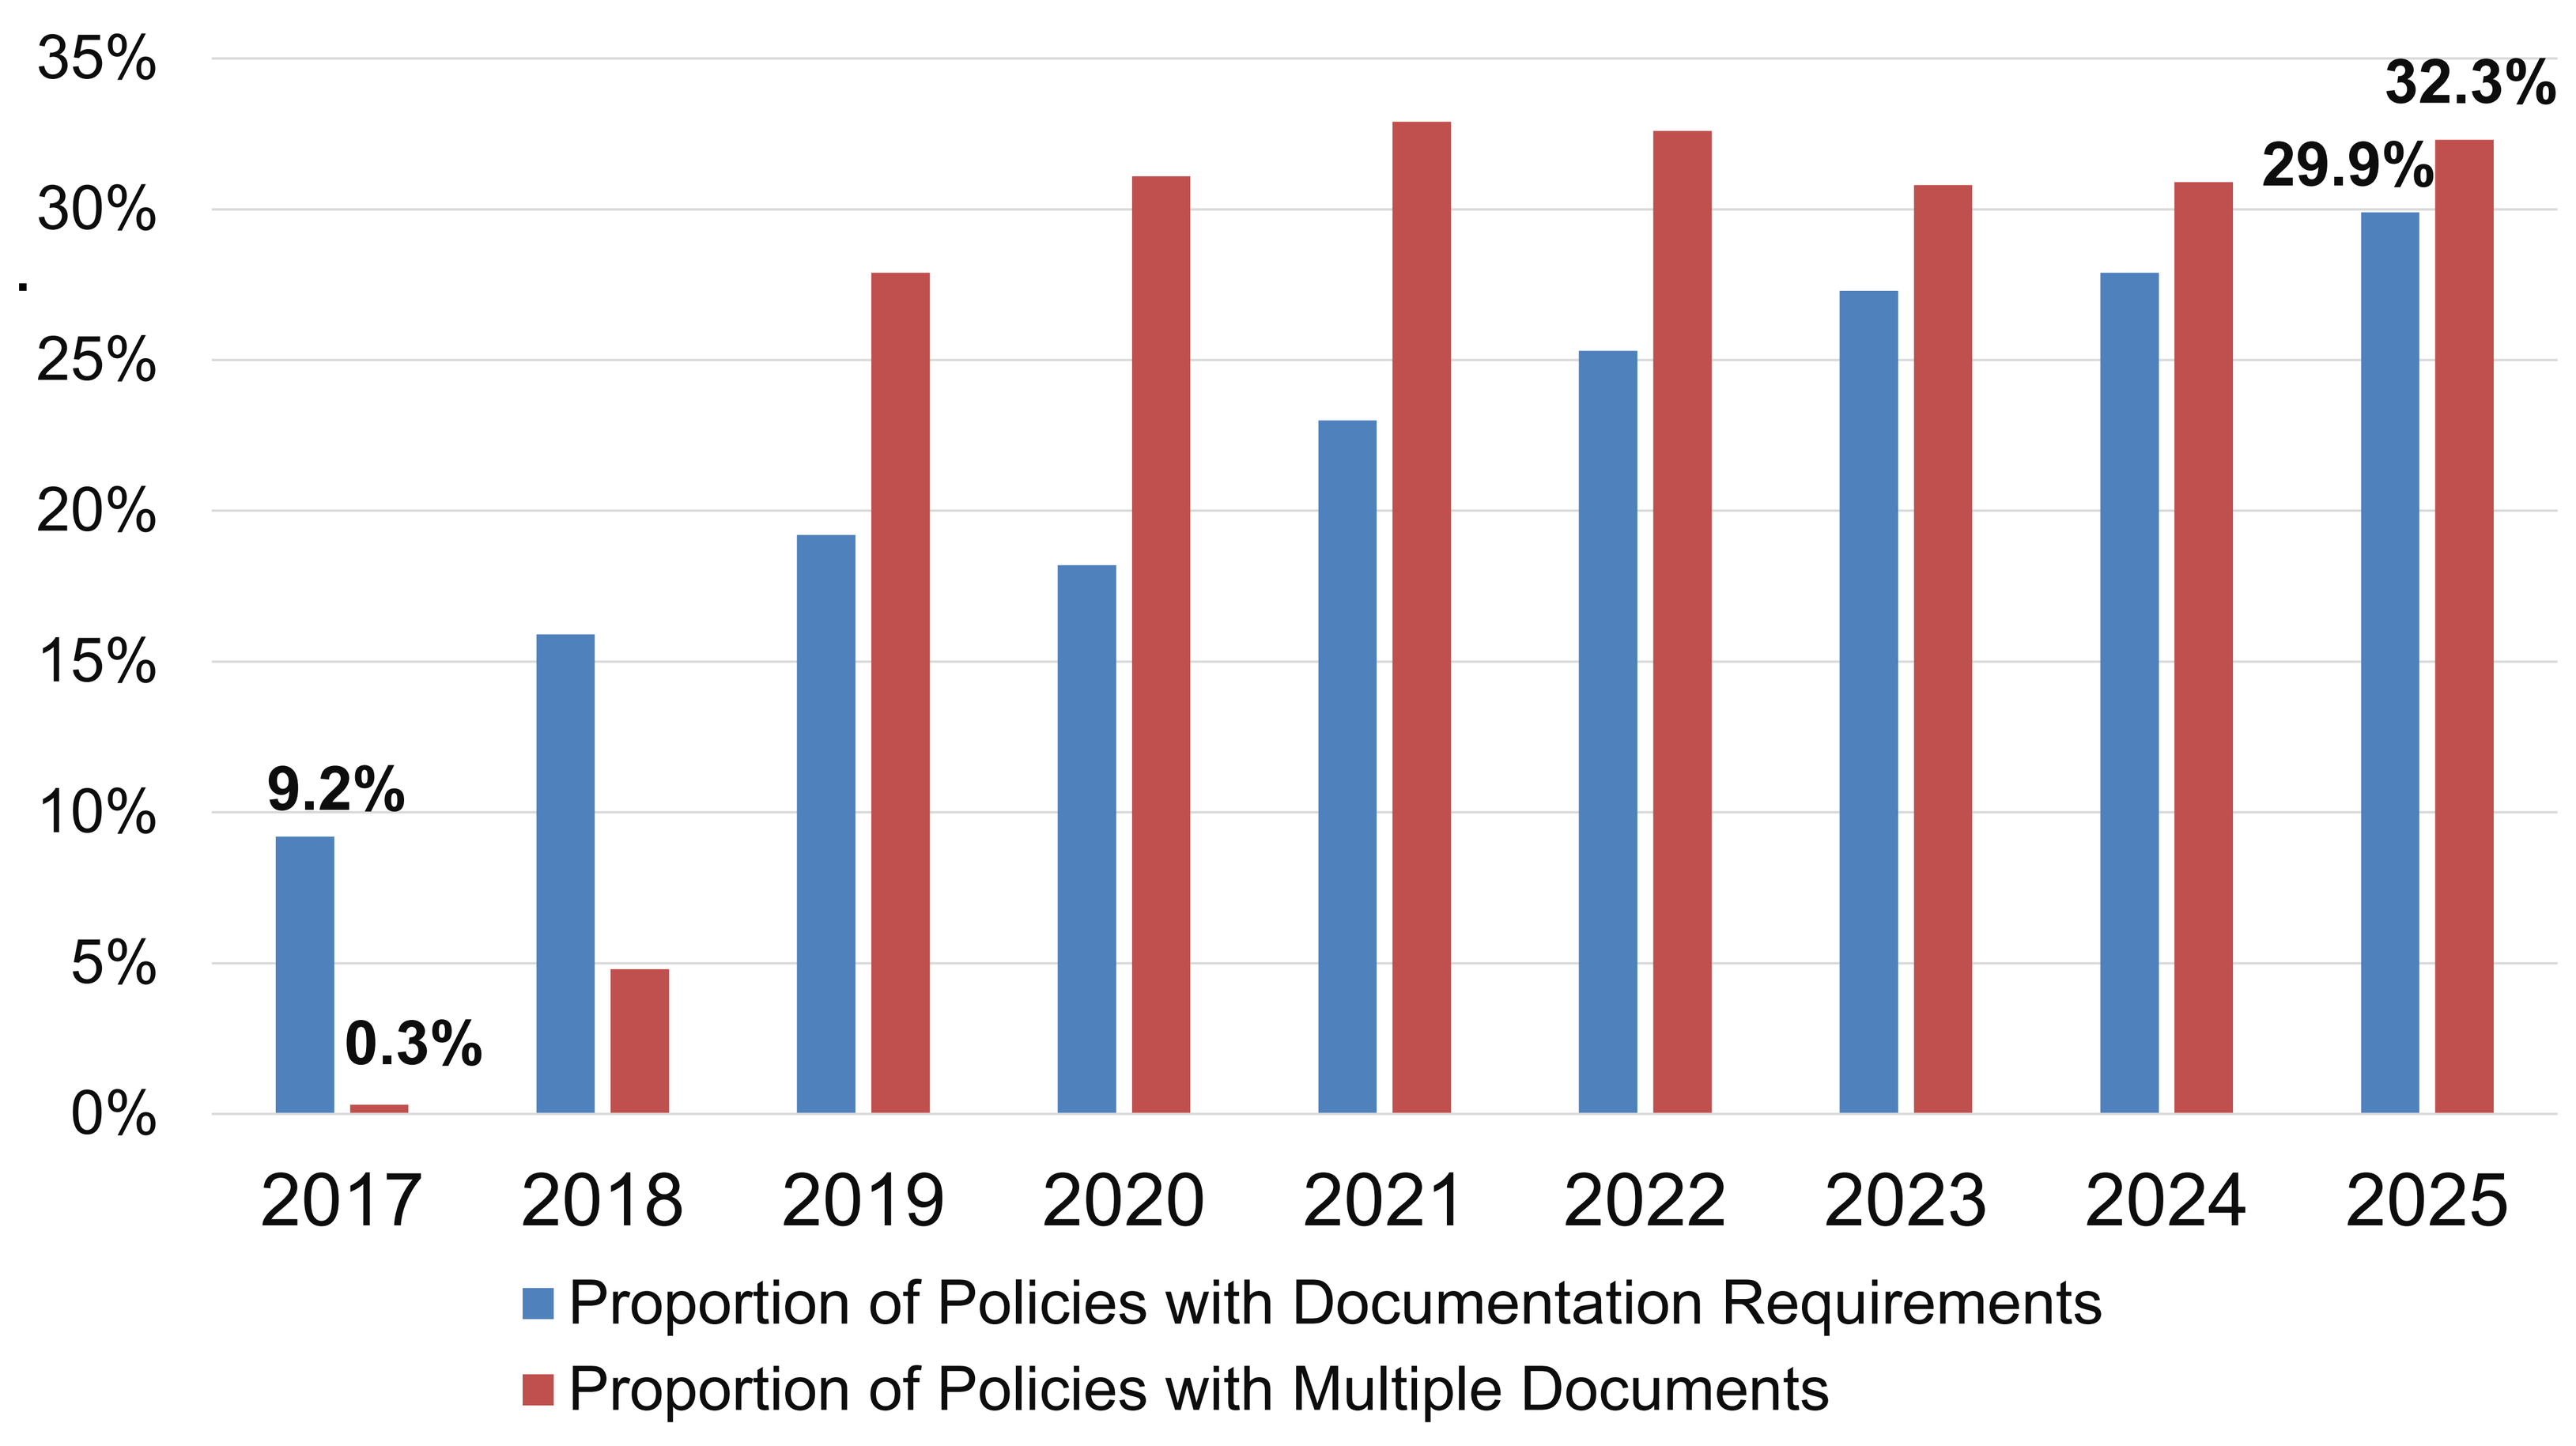
35%
30%
25%
20%
15%
10%
5%
0%
9.2%
0.3%
29.9%
32.3%
2017 2018 2019 2020 2021 2022 2023 2024 2025
Proportion of Policies with Documentation Requirements
Proportion of Policies with Multiple Documents

- In 2025, the following measures of coverage policy complexity varied by health plan:
 - Mean word count (119.3 – 922.4 words)
 - Number of coverage criteria (8.8 – 48.7 criteria)
 - Proportion of policies with multiple documents (0% - 93.5%)
 - Proportion of policies with pre-authorization documentation requirements (3.9% - 96.7%)

Figure 3. Variation in Specialty Drug Coverage Policy Length, by Health Plan (2025)

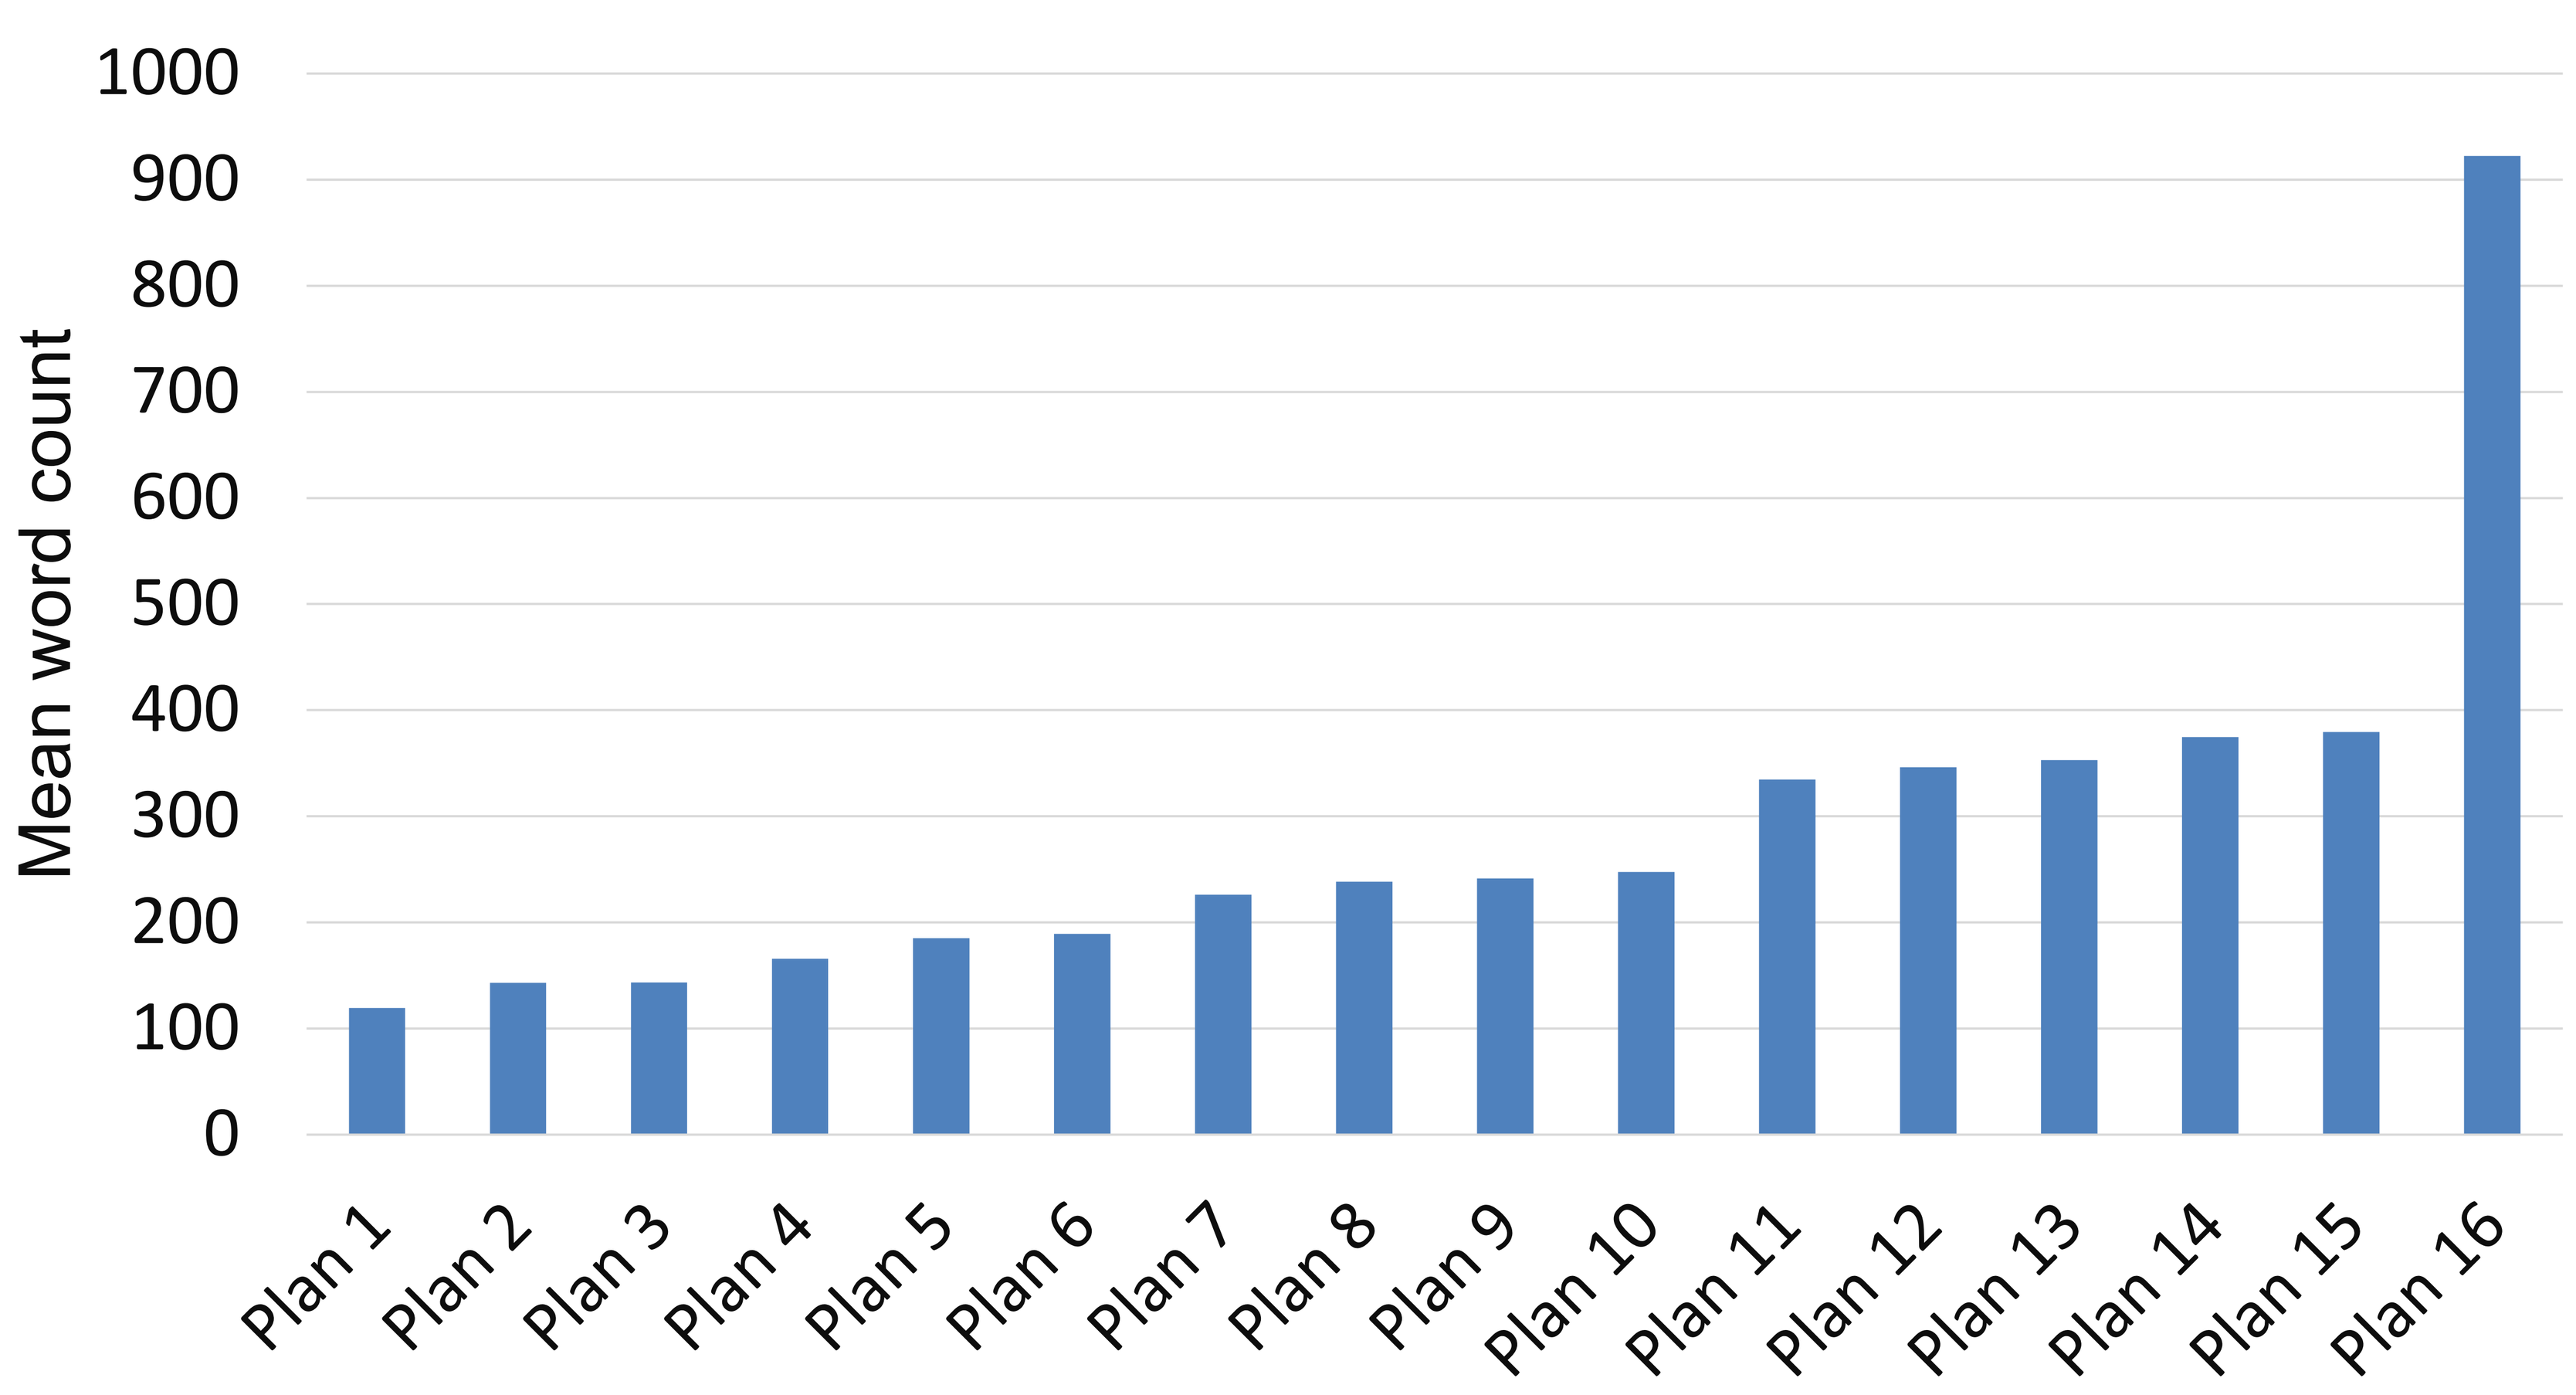
Mean word count
1000
900
800
700
600
500
400
300
200
100
0
Plan 1 Plan 2 Plan 3 Plan 4 Plan 5 Plan 6 Plan 7 Plan 8 Plan 9 Plan 10 Plan 11 Plan 12 Plan 13 Plan 14 Plan 15 Plan 16

Figure 4. Variation in Documentation Requirements, by Health Plan (2025)

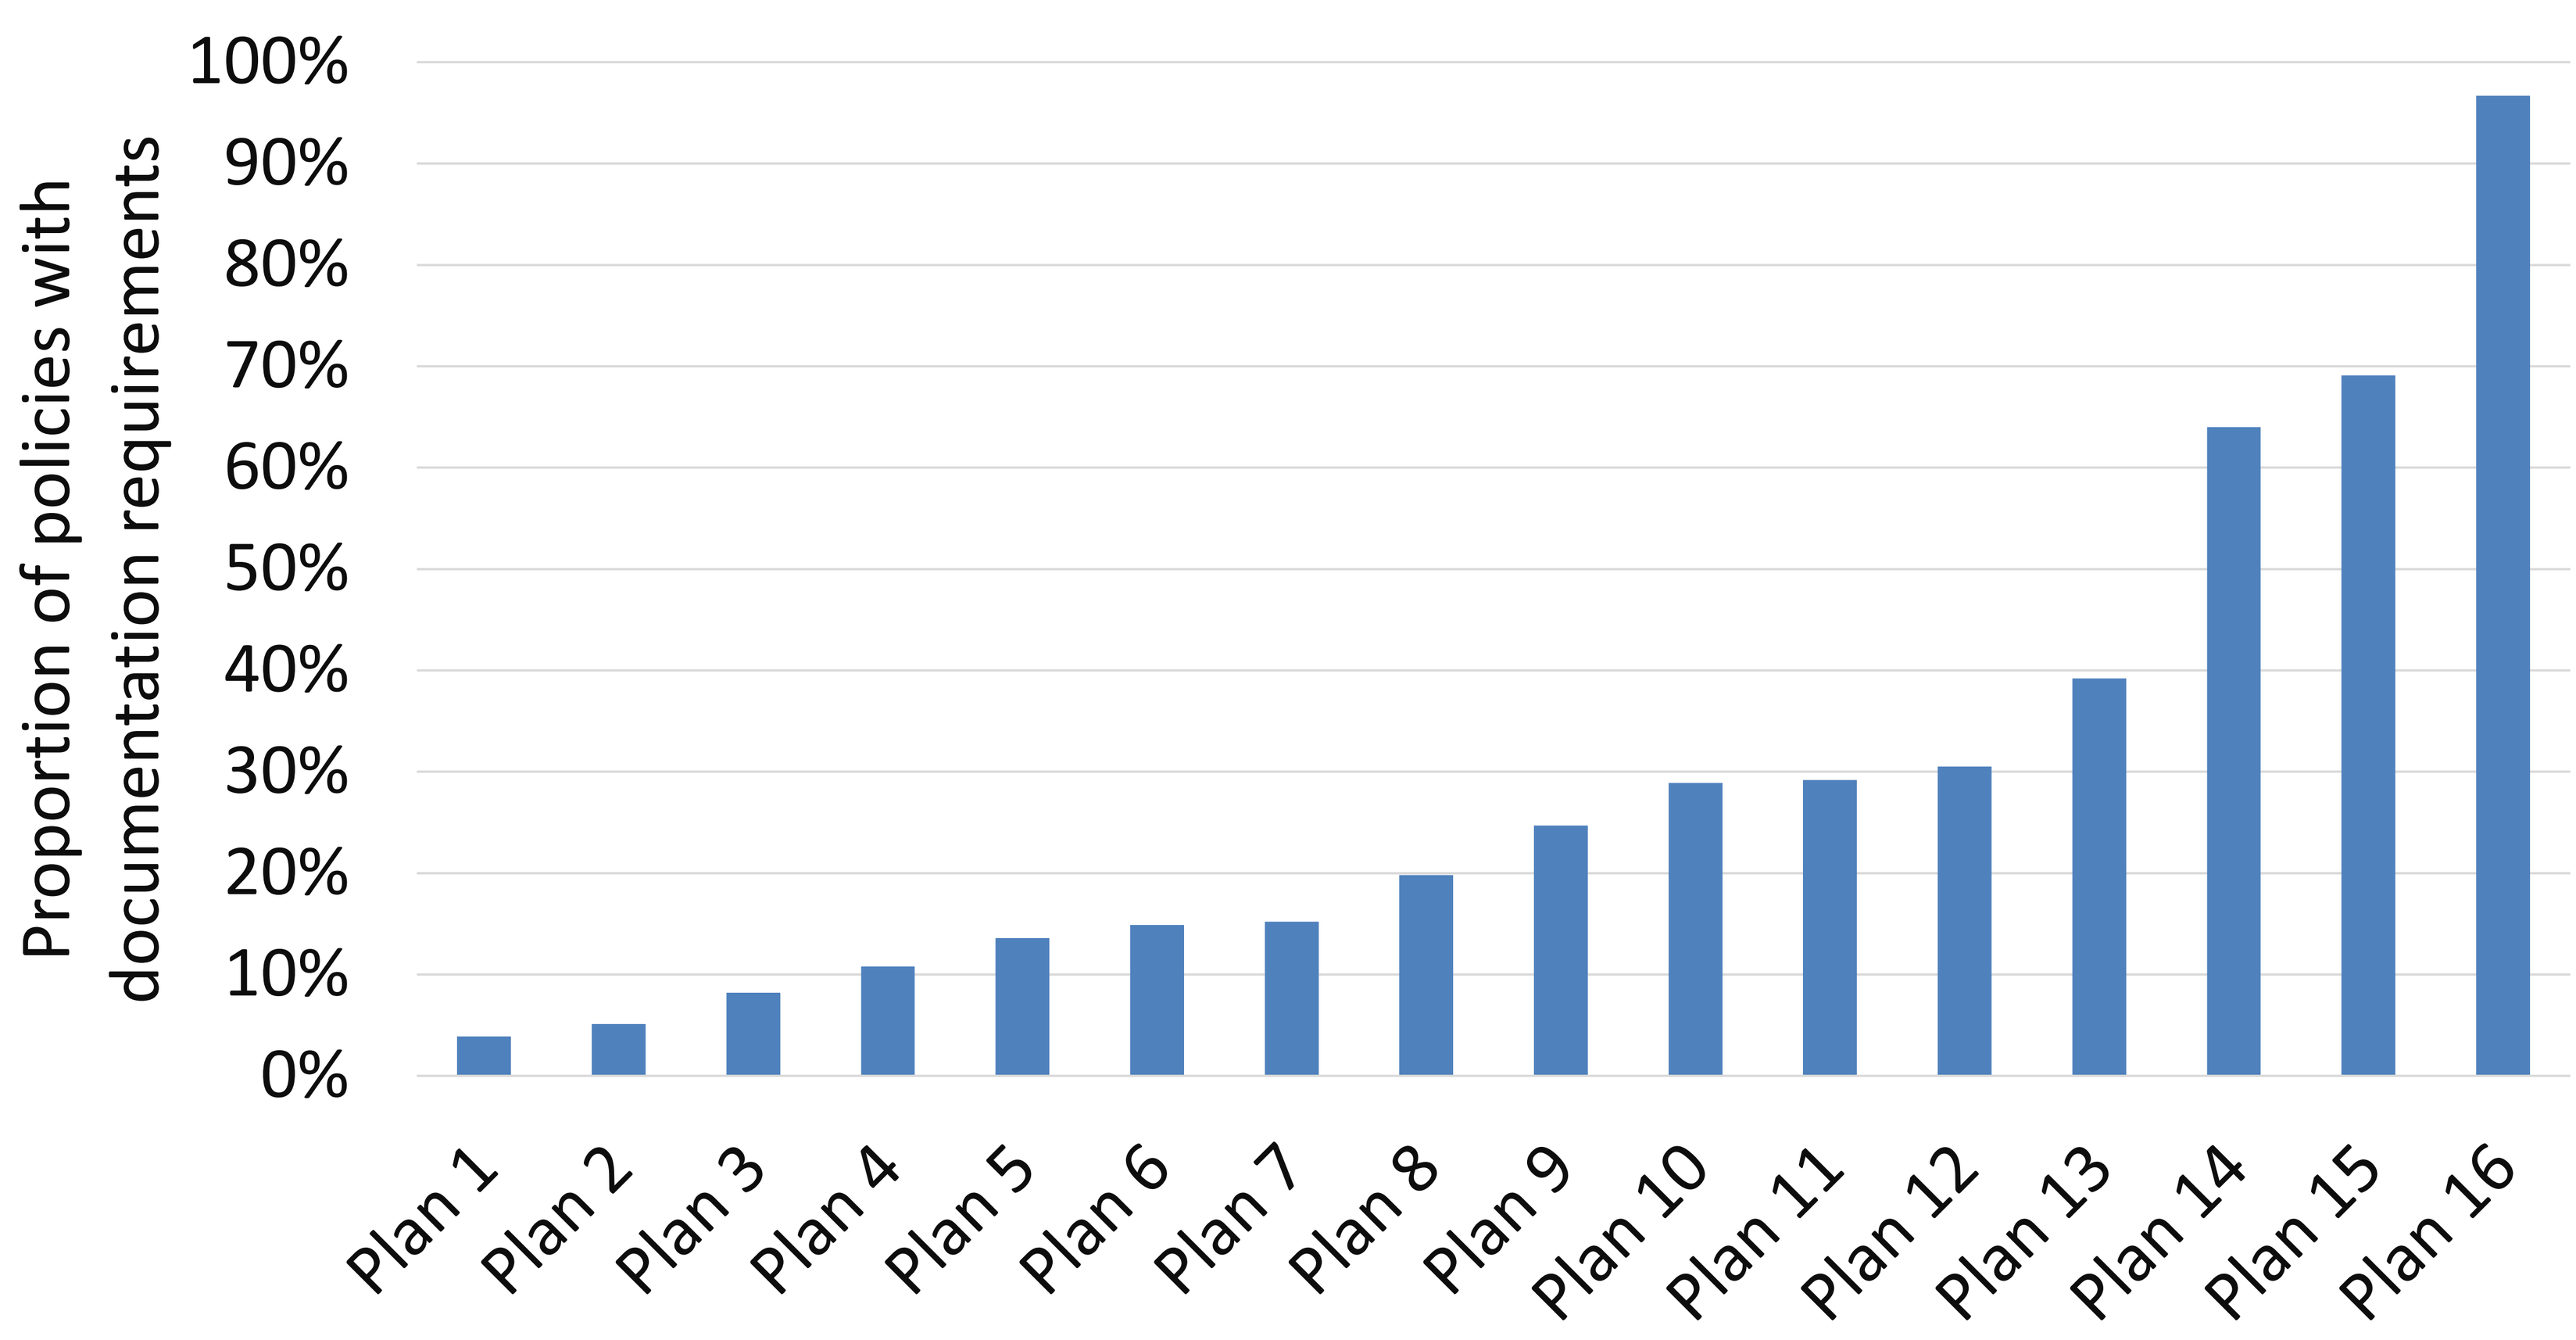
Proportion of policies with documentation requirements
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
Plan 1 Plan 2 Plan 3 Plan 4 Plan 5 Plan 6 Plan 7 Plan 8 Plan 9 Plan 10 Plan 11 Plan 12 Plan 13 Plan 14 Plan 15 Plan 16

***Health plans are ordered by magnitude, plan 1 in Figure 3 varies from Figure 4.**

CONCLUSION

- Specialty drug coverage policies have become increasingly complex over time.
- Health plans now issue more documents per policy, use longer coverage language with a greater number of specific criteria, and are more likely to require pre-authorization documentation requirements.
- Greater complexity may delay prescribing processes, increase administrative demands on providers, and delay patient treatment initiation.
- Future research is needed to evaluate how policy complexity affects real-world drug utilization and patient outcomes.

REFERENCES

1. Howell S, et al. Quantifying The Economic Burden Of Drug Utilization Management On Payers, Manufacturers, Physicians, And Patients. Health Aff (Millwood). 2021 Aug;40(8):1206-1214
2. Edwards ML, Yin PT, Kuehn M, et al. Physician perceptions of drug utilization management: Results of a national survey. *PLoS One*. 2022;17(9):e0274772.

FUNDING

This research was funded by the Pharmaceutical Research and Manufacturers of America (PHRMA)

CONTACT

For more information about the SPEC Database, please contact James Chambers at james.chambers@tuftsmedicine.org.

Poster Code: HSD99